Clinical Development of Inhaled Interferon Beta-1a (SNG001) for the Treatment of Patients with COVID-19 in the Home and Hospital Settings

P. Monk[1], J. Brookes[1], V. Tear[1], T. Batten[2], M. Mankowski[3], F. Gabbay[3], G. Walters[1], D. Davies[4], C. Brightling[5], M. Kraft[6], R. Djukanovic[4], S. Holgate[4] & T. Wilkinson[4]

[1] Synairgen Research Ltd., UK, [2]Veramed, UK, [3]TranScrip, UK, [4]University of Southampton, UK, [5]University of Leicester, UK, [6]Mount Sinai Medical Center, New York, USA.

Inhaled Interferon beta (IFN-β) for Severe Viral Lung Infections

- IFN-β is a naturally occurring protein which orchestrates the body's antiviral defences
- Type I IFN deficiency, due to factors like genetic make up, disease or an aging immune system, is associated with severe viral lower respiratory tract illness
- Viruses suppress IFN-β production by cells to evade the host immune response
- IFN-β shows antiviral activity in cell-based assays against a broad range of viruses
- The aim of the inhaled route of delivery is to restore IFN-β levels at the site of infection in the lungs, suppressed by the virus or reduced due to host factors, to drive antiviral responses to clear the virus and to minimise systemic exposure.

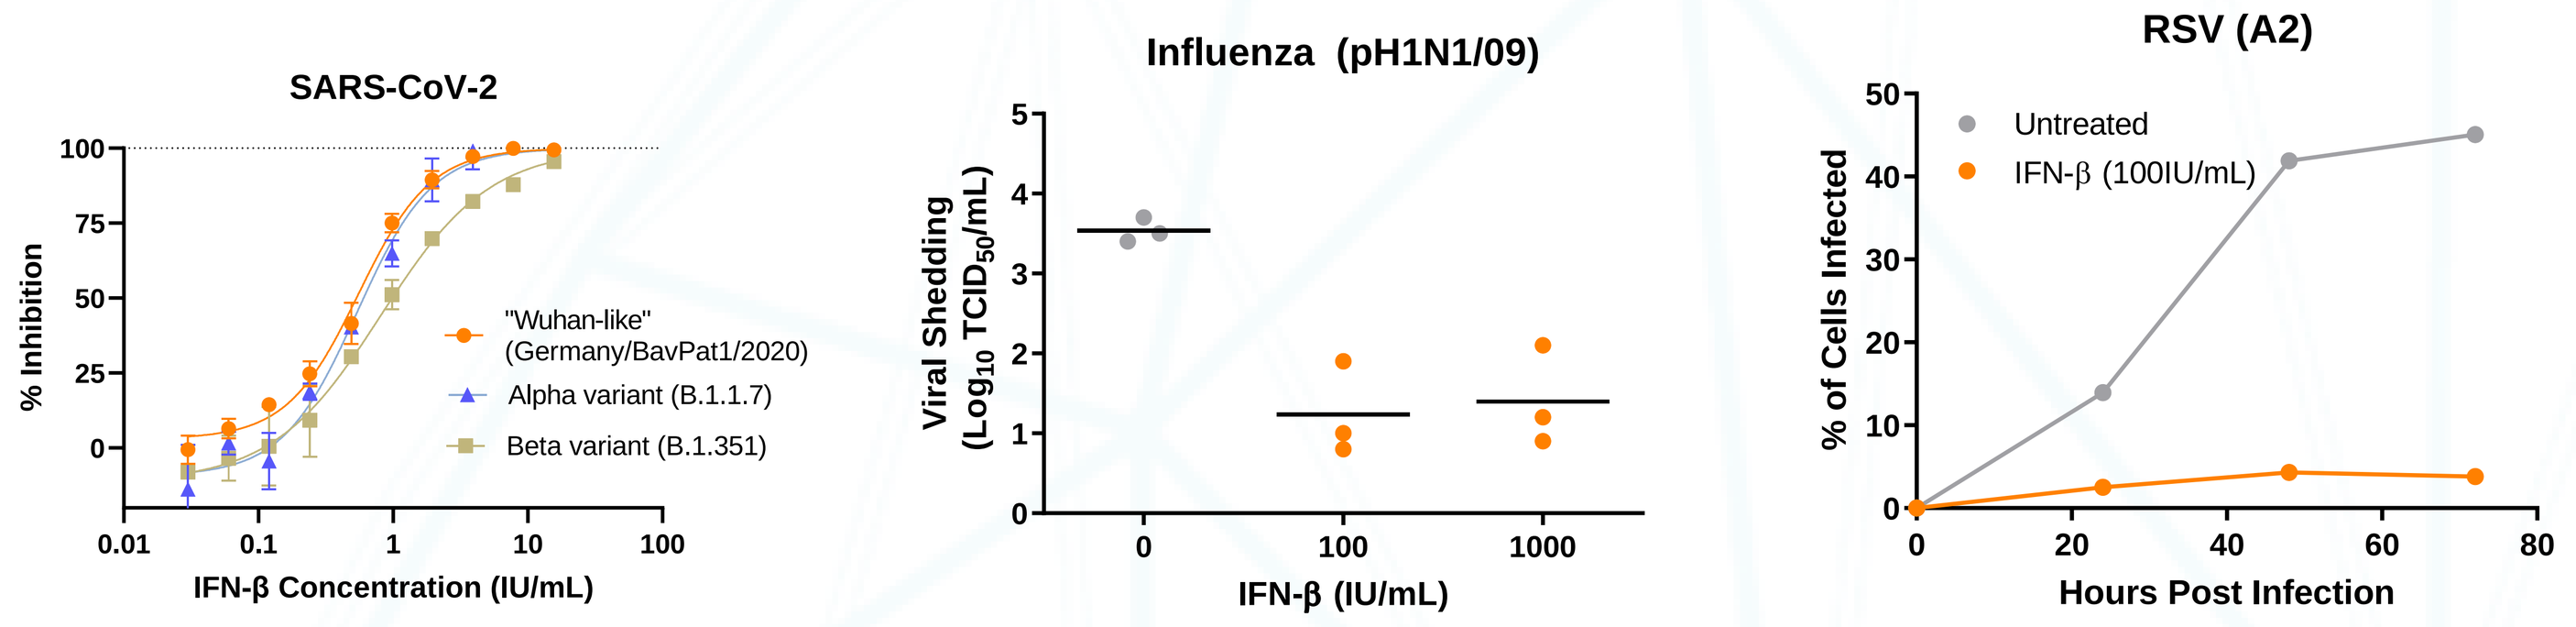
SARS-CoV-2
% Inhibition
"Wuhan-like" (Germany/BavPat1/2020)
Alpha variant (B.1.1.7)
Beta variant (B.1.351)
IFN-β Concentration (IU/mL)
Influenza (pH1N1/09)
Viral Shedding (Log10 TCID50/mL)
IFN-β (IU/mL)
RSV (A2)
% of Cells Infected
Untreated
IFN-β (100IU/mL)
Hours Post Infection

Antiviral activity of SNG001 (IFN-β) in cell based assays

Summary of Clinical Development of SNG001 in COVID-19

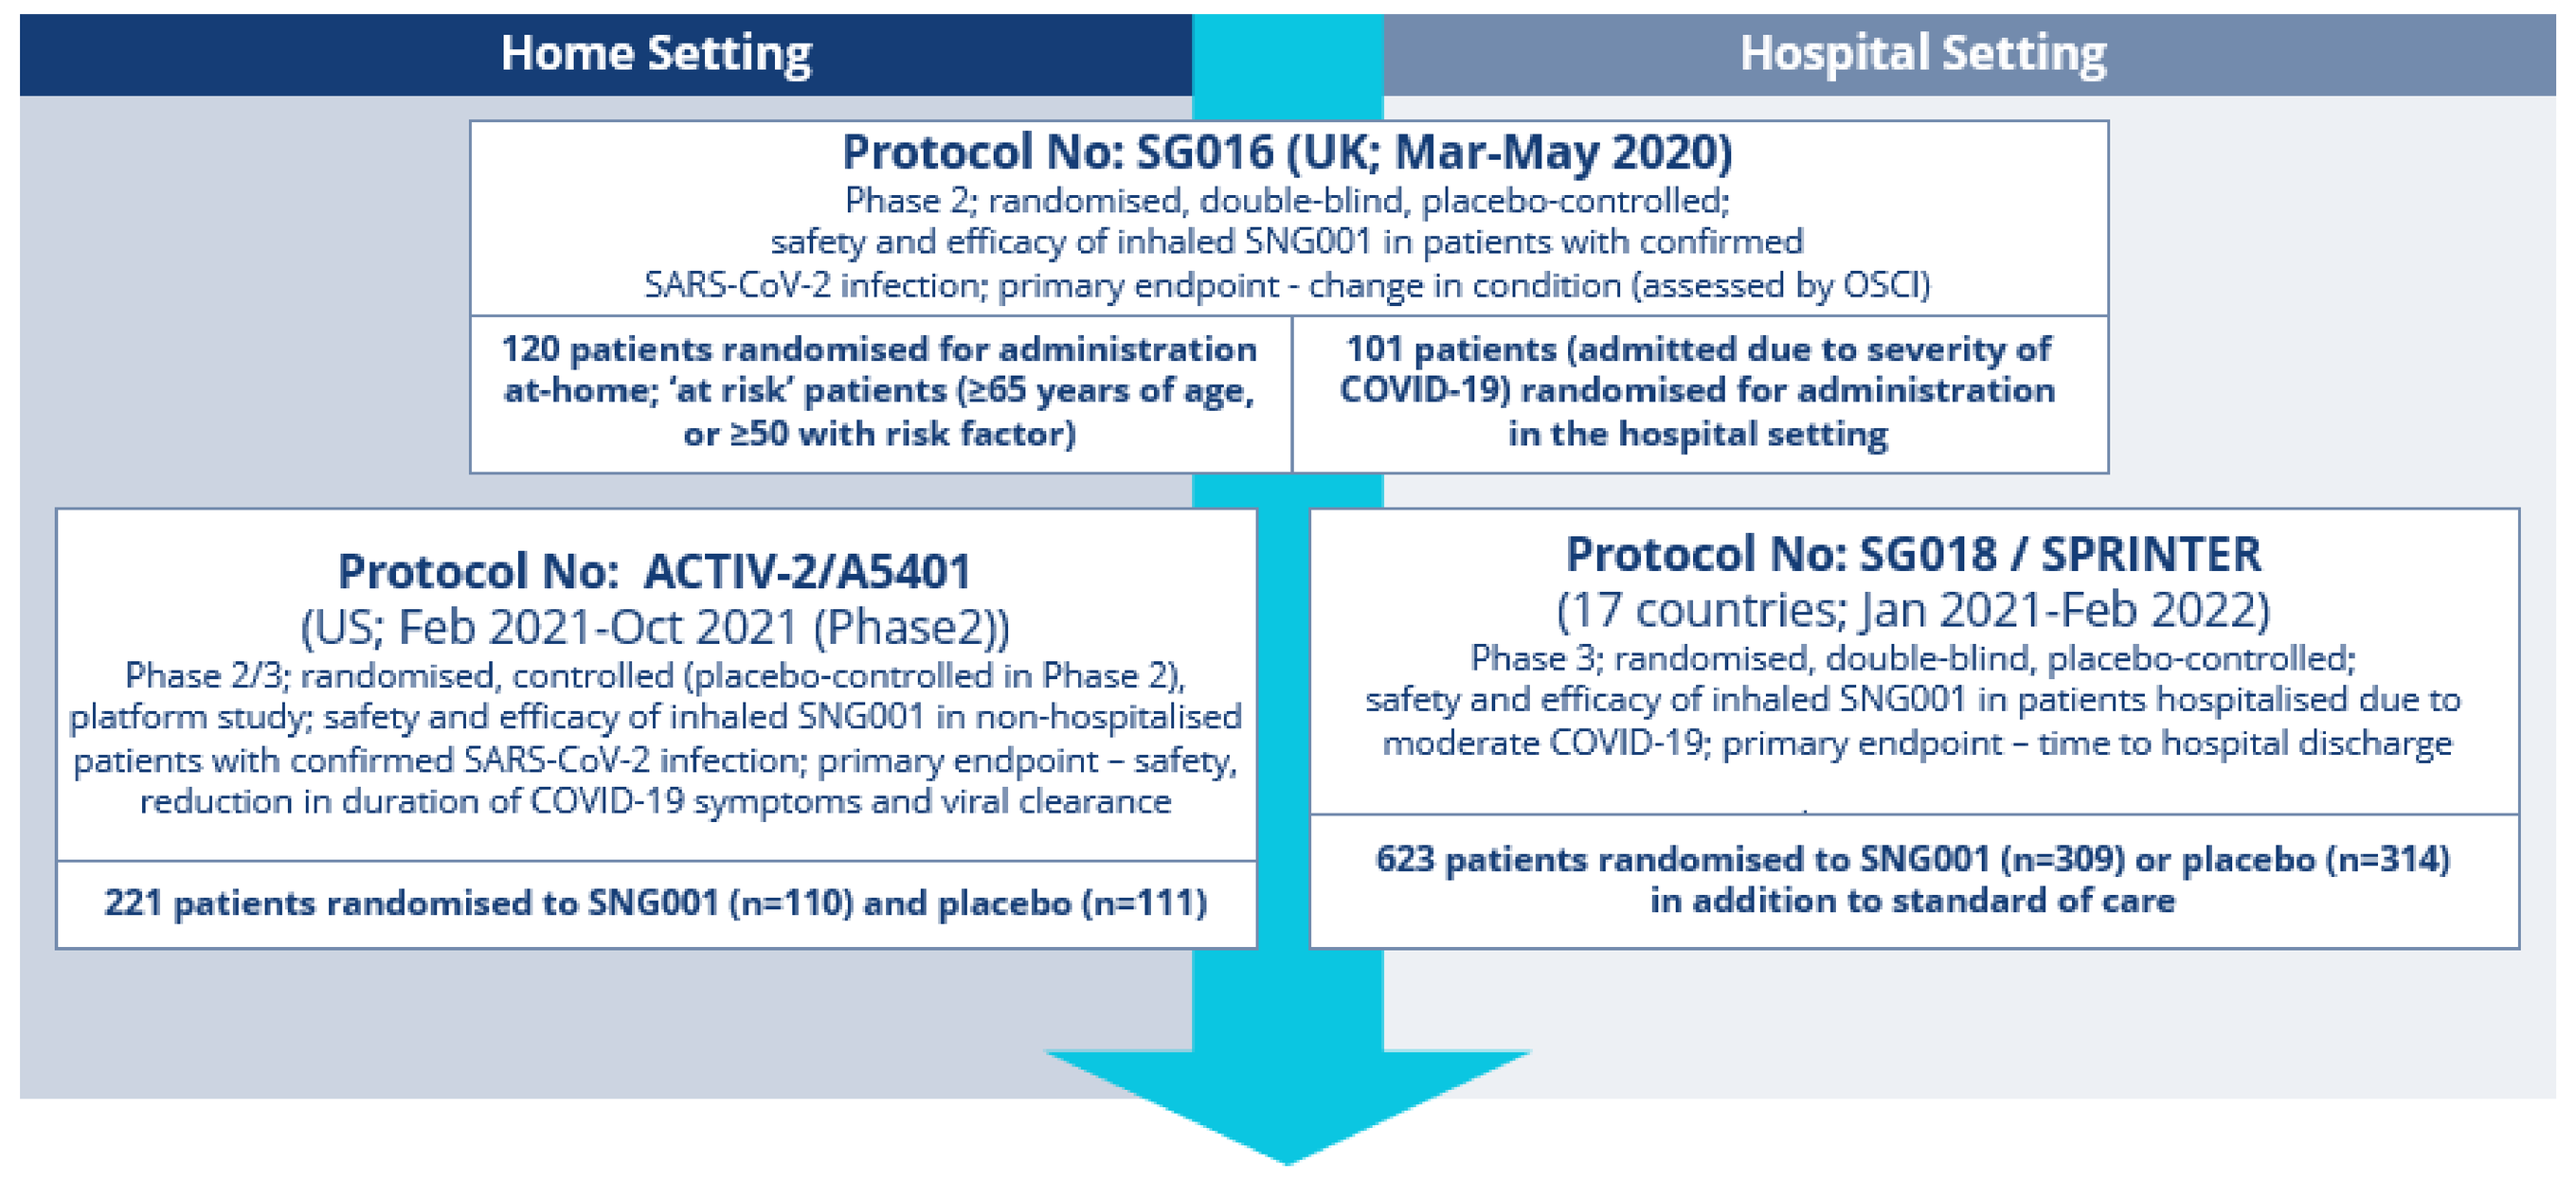
Home Setting
Hospital Setting
Protocol No: SG016 (UK; Mar-May 2020)
Phase 2; randomised, double-blind, placebo-controlled; safety and efficacy of inhaled SNG001 in patients with confirmed SARS-CoV-2 infection; primary endpoint - change in condition (assessed by OSCI)
120 patients randomised for administration at-home; 'at risk' patients (≥65 years of age, or ≥50 with risk factor)
101 patients (admitted due to severity of COVID-19) randomised for administration in the hospital setting
Protocol No: ACTIV-2/A5401
(US; Feb 2021-Oct 2021 (Phase2))
Phase 2/3; randomised, controlled (placebo-controlled in Phase 2), platform study; safety and efficacy of inhaled SNG001 in non-hospitalised patients with confirmed SARS-CoV-2 infection; primary endpoint – safety, reduction in duration of COVID-19 symptoms and viral clearance
221 patients randomised to SNG001 (n=110) and placebo (n=111)
Protocol No: SG018 / SPRINTER
(17 countries; Jan 2021-Feb 2022)
Phase 3; randomised, double-blind, placebo-controlled; safety and efficacy of inhaled SNG001 in patients hospitalised due to moderate COVID-19; primary endpoint – time to hospital discharge
623 patients randomised to SNG001 (n=309) or placebo (n=314) in addition to standard of care

Substantial Clinical Safety Database from 7 Clinical Trials (Home and Hospital)

- A total of 750 patients have been treated with SNG001, the majority of which had a confirmed or suspected respiratory viral infection (rhinovirus, influenza, SARS-CoV-2, etc)
- 515 patients had COVID-19; 67 had COPD; and 168 patients had asthma
- Favourable safety profile – generally fewer patients on SNG001 reporting any adverse event or any serious adverse event versus patients on placebo. No specific safety signals were seen.
- Well-tolerated locally (lung) – lack of significant systemic tolerability concerns consistent with local delivery and negligible systemic exposure (or ADAs) to SNG001 after inhalation

Clinical Trial Findings Support Further Clinical Development

- There remains a strong clinical rationale for developing inhaled IFN-β as a broad-spectrum antiviral for severe viral lung infections. Clinical trial results including favourable safety profile, support continued investigation of SNG001 in trials in COVID-19 and against seasonal cold and flu viruses evaluating:
 - Prevention of hospitalisation in at risk patients in the home setting
 - Prevention of progression and/or mortality in at risk hospitalised patients
 - Prevention of the development of post viral symptoms (data on long COVID symptoms presented at ID Week 2022)

SG016 Trial Design (Hospital Setting)

Study Title	A randomised, double-blind, placebo-controlled trial to determine the safety and efficacy of inhaled SNG001 (IFN-β1a for nebulisation) for the treatment of patients with confirmed SARS-CoV-2 infection (NCT04385095)
Primary Endpoint	Change in clinical condition on the WHO Ordinal Scale for Clinical Improvement (OSCI) during the dosing period
Setting	Symptomatic, hospitalised adults with COVID-19
Number of Patients	101 patients were randomly assigned to SNG001 (n=50) or placebo (n=51). 48 received SNG001 and 50 received placebo and were included in the ITT
Dosing Regimen	SNG001(6 MIU interferon beta-1a) or placebo (randomised 1:1) delivered by I-neb nebuliser with a filter on the exhalation port, once-a-day for up to 14 days on top of standard of care

SG016 Results

- The graph below shows recovery to 'no limitation of activity' across the 14 day treatment period. Patients treated with SNG001 were more than twice as likely to recover compared to those receiving placebo (p=0.043).
- Whilst not powered to show it there were also trends towards prevention of progression to severe disease and faster hospital discharge (see Table below).
- SNG001 was well-tolerated and had a favourable safety profile.
- These data from the Phase 2 study supported progression to the Phase 3 SPRINTER trial.

Analysis of the WHO OSCI during the dosing period

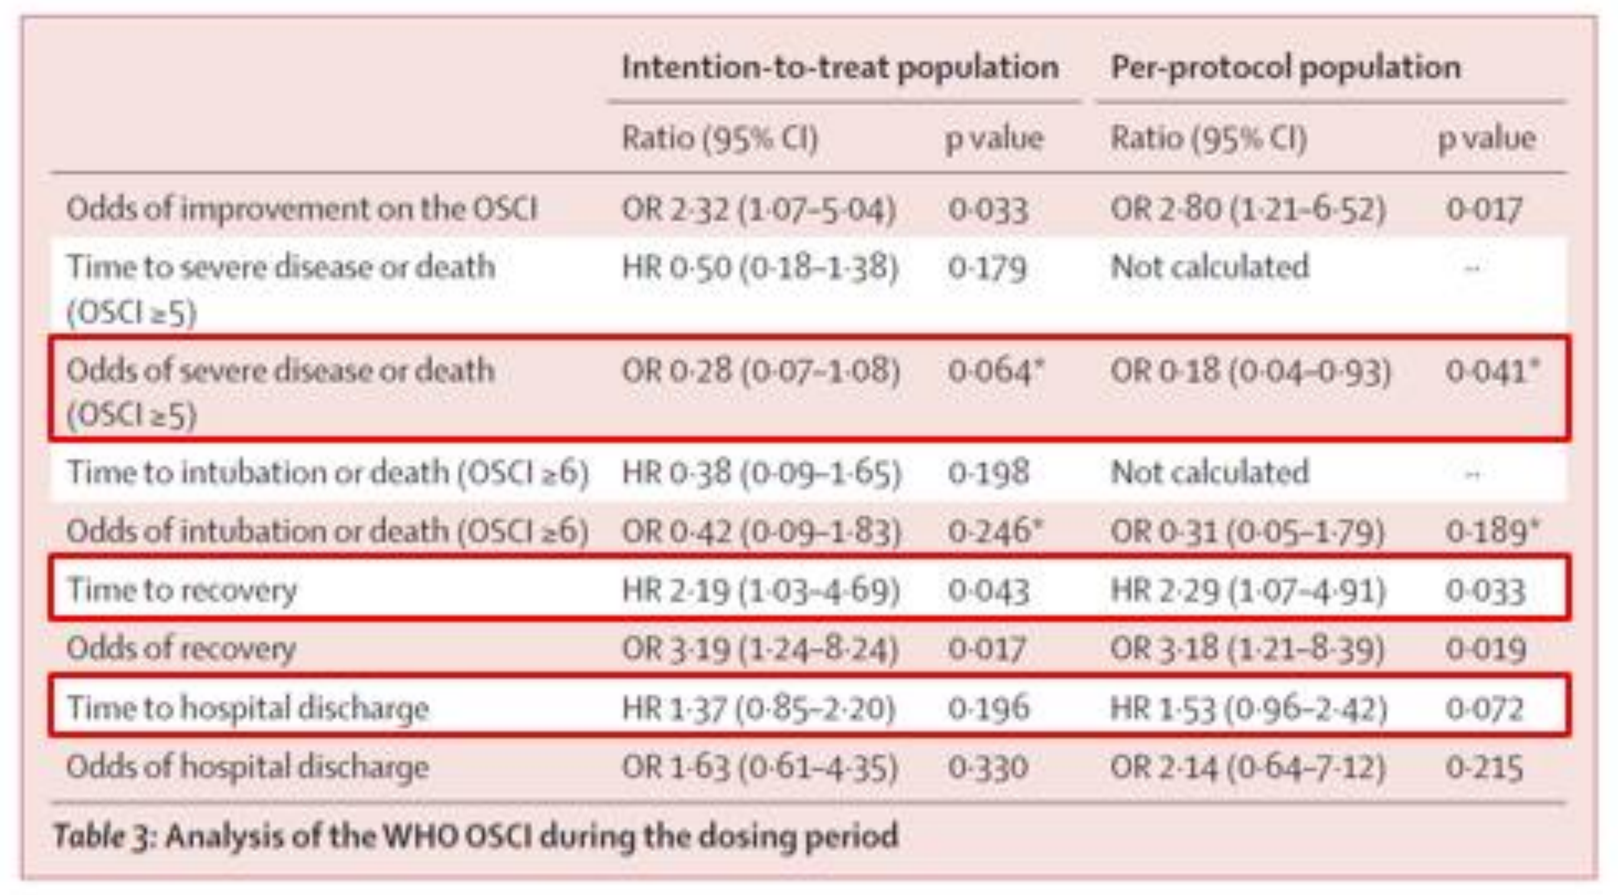

	Intention-to-treat population		Per-protocol population	
	Ratio (95% CI)	p value	Ratio (95% CI)	p value
Odds of improvement on the OSCI	OR 2·32 (1·07–5·04)	0·033	OR 2·80 (1·21–6·52)	0·017
Time to severe disease or death (OSCI ≥5)	HR 0·50 (0·18–1·38)	0·179	Not calculated	..
Odds of severe disease or death (OSCI ≥5)	OR 0·28 (0·07–1·08)	0·064*	OR 0·18 (0·04–0·93)	0·041*
Time to intubation or death (OSCI ≥6)	HR 0·38 (0·09–1·65)	0·198	Not calculated	..
Odds of intubation or death (OSCI ≥6)	OR 0·42 (0·09–1·83)	0·246*	OR 0·31 (0·05–1·79)	0·189*
Time to recovery	HR 2·19 (1·03–4·69)	0·043	HR 2·29 (1·07–4·91)	0·033
Odds of recovery	OR 3·19 (1·24–8·24)	0·017	OR 3·18 (1·21–8·39)	0·019
Time to hospital discharge	HR 1·37 (0·85–2·20)	0·196	HR 1·53 (0·96–2·42)	0·072
Odds of hospital discharge	OR 1·63 (0·61–4·35)	0·330	OR 2·14 (0·64–7·12)	0·215

Table 3: Analysis of the WHO OSCI during the dosing period

Recovery (intention-to-treat population)

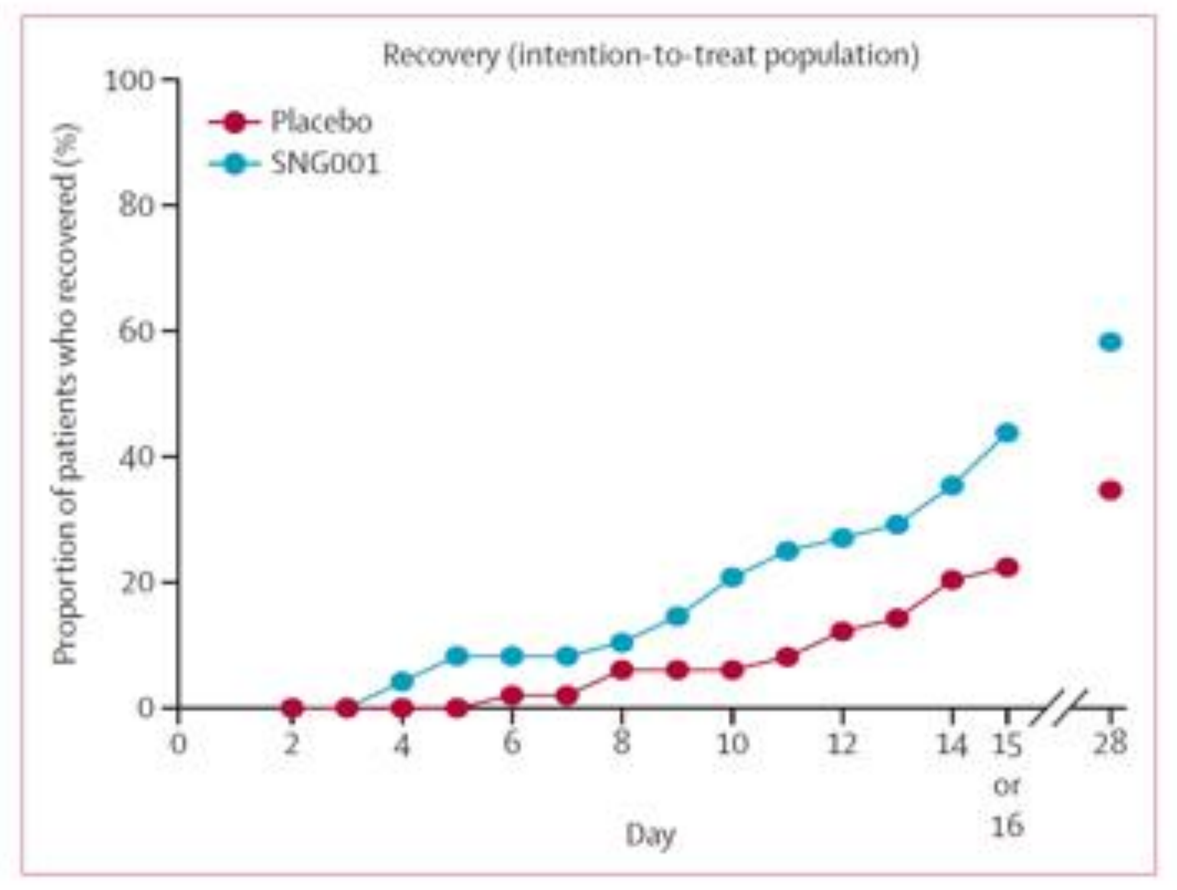
Recovery (intention-to-treat population)
Placebo
SNG001
Proportion of patients who recovered (%)
Day
15 or 16

Monk *et al.* (2021) Lancet Respir Med; 9(2):196-206

SG018 (SPRINTER) Trial Design (Hospital Setting)

Study Title	A randomised, double-blind, placebo-controlled, Phase 3 trial to determine the efficacy and safety of inhaled SNG001 for the treatment of patients hospitalised due to moderate COVID-19 (NCT04732949)
Primary Endpoints	Time to hospital discharge Time to recovery to 'No Limitation of Activities' up to Day 28
Key Secondary Endpoints	Progression to severe disease or death within 35 days Progression to intubation or death within 35 days Death within 35 days
Setting	Patients hospitalised due to COVID-19 requiring supplemental oxygen by mask or nasal prongs
Number of Patients	623 patients randomised to SNG001 (n=309) or placebo (n=314) at 100+ sites in Europe, North America, South America, India, Israel
Dosing Regimen	SNG001(15.6 MIU interferon beta-1a) or placebo (randomised 1:1) delivered by Aerogen Solo/Ultra vibrating mesh nebuliser with a filter on the exhalation port, once-a-day for up to 14 days on top of standard of care

SG018 (SPRINTER) Results

- The study did not meet its primary efficacy endpoints of hospital discharge and recovery, most likely due to improvement in SoC since we conducted the Phase 2 study at the start of the pandemic, which may have reduced the window to show a treatment effect.
- There was an encouraging signal for the key secondary endpoint of progression to severe disease or death (26% RRR in ITT, 36% RRR in PP*).
- *Post hoc* analyses in at risk groups showed stronger treatment effects increasing confidence SNG001 was having a beneficial effect on disease progression.
- SNG001 was well-tolerated and had a favourable safety profile.

Progression to severe disease or death

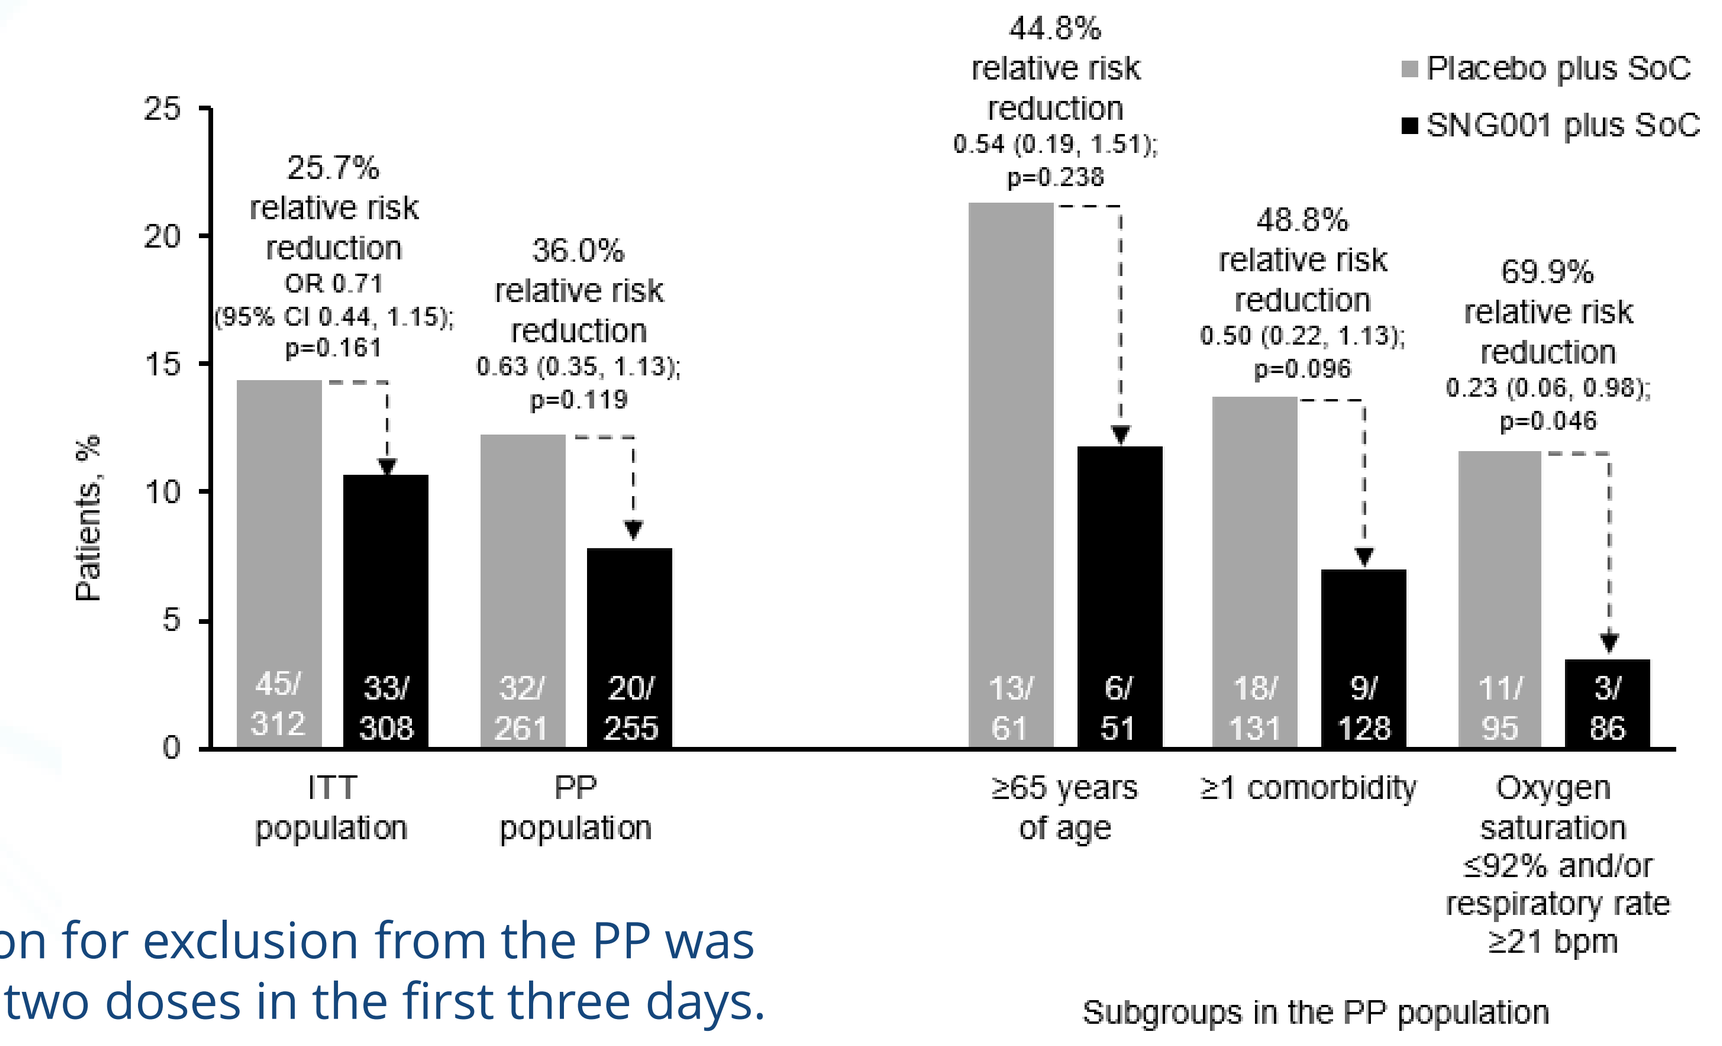
Patients, %
25.7% relative risk reduction OR 0.71 (95% CI 0.44, 1.15); p=0.161
36.0% relative risk reduction 0.63 (0.35, 1.13); p=0.119
44.8% relative risk reduction 0.54 (0.19, 1.51); p=0.238
48.8% relative risk reduction 0.50 (0.22, 1.13); p=0.096
69.9% relative risk reduction 0.23 (0.06, 0.98); p=0.046
Placebo plus SoC
SNG001 plus SoC
45/312
33/308
32/261
20/255
13/61
6/51
18/131
9/128
11/95
3/86
ITT population
PP population
≥65 years of age
≥1 comorbidity
Oxygen saturation ≤92% and/or respiratory rate ≥21 bpm
Subgroups in the PP population

*The main reason for exclusion from the PP was a failure to take two doses in the first three days.

Monk *et al.* (2023) ERJ Open Res. 2023;9(2):00605-2022.

ACTIV-2 (A5401) Trial Design (Home Setting)

Study Title	Phase 2/3 Adaptive Platform Treatment Trial for Outpatients with COVID-19 (Adapt Out COVID) (NCT04518410)
Primary Endpoints	Safety Change in clinical condition on the WHO Ordinal Scale for Clinical Improvement (OSCI) during the dosing period
Setting	Symptomatic, non-hospitalised adults with COVID-19
Number of Patients	110 participants per investigational agent (and 110 on placebo) recruited across 25 US sites
Dosing Regimen	SNG001(15.6 MIU interferon beta-1a) or placebo (randomised 1:1) delivered by Aerogen Solo/Ultra vibrating mesh nebuliser with a filter on the exhalation port, once-a-day for up to 14 days on top of standard of care

ACTIV-2 (A5401) Results

- There were no statistically significant differences with respect to safety, symptom resolution, or virology.
- Fewer participants treated with SNG001 were hospitalised
 - 86% RRR (p=0.07; Two-sided Fisher's exact test)
 - There were no deaths in either arm
 - In the SG016 Synairgen Phase 2 study (Home setting), 2/58 (3.4%) placebo patients versus 0/56 (0%) SNG001 patients were hospitalised
- SNG001 was well-tolerated and had a favourable safety profile
- DSMB recommended progression to Phase 3 in October 2021

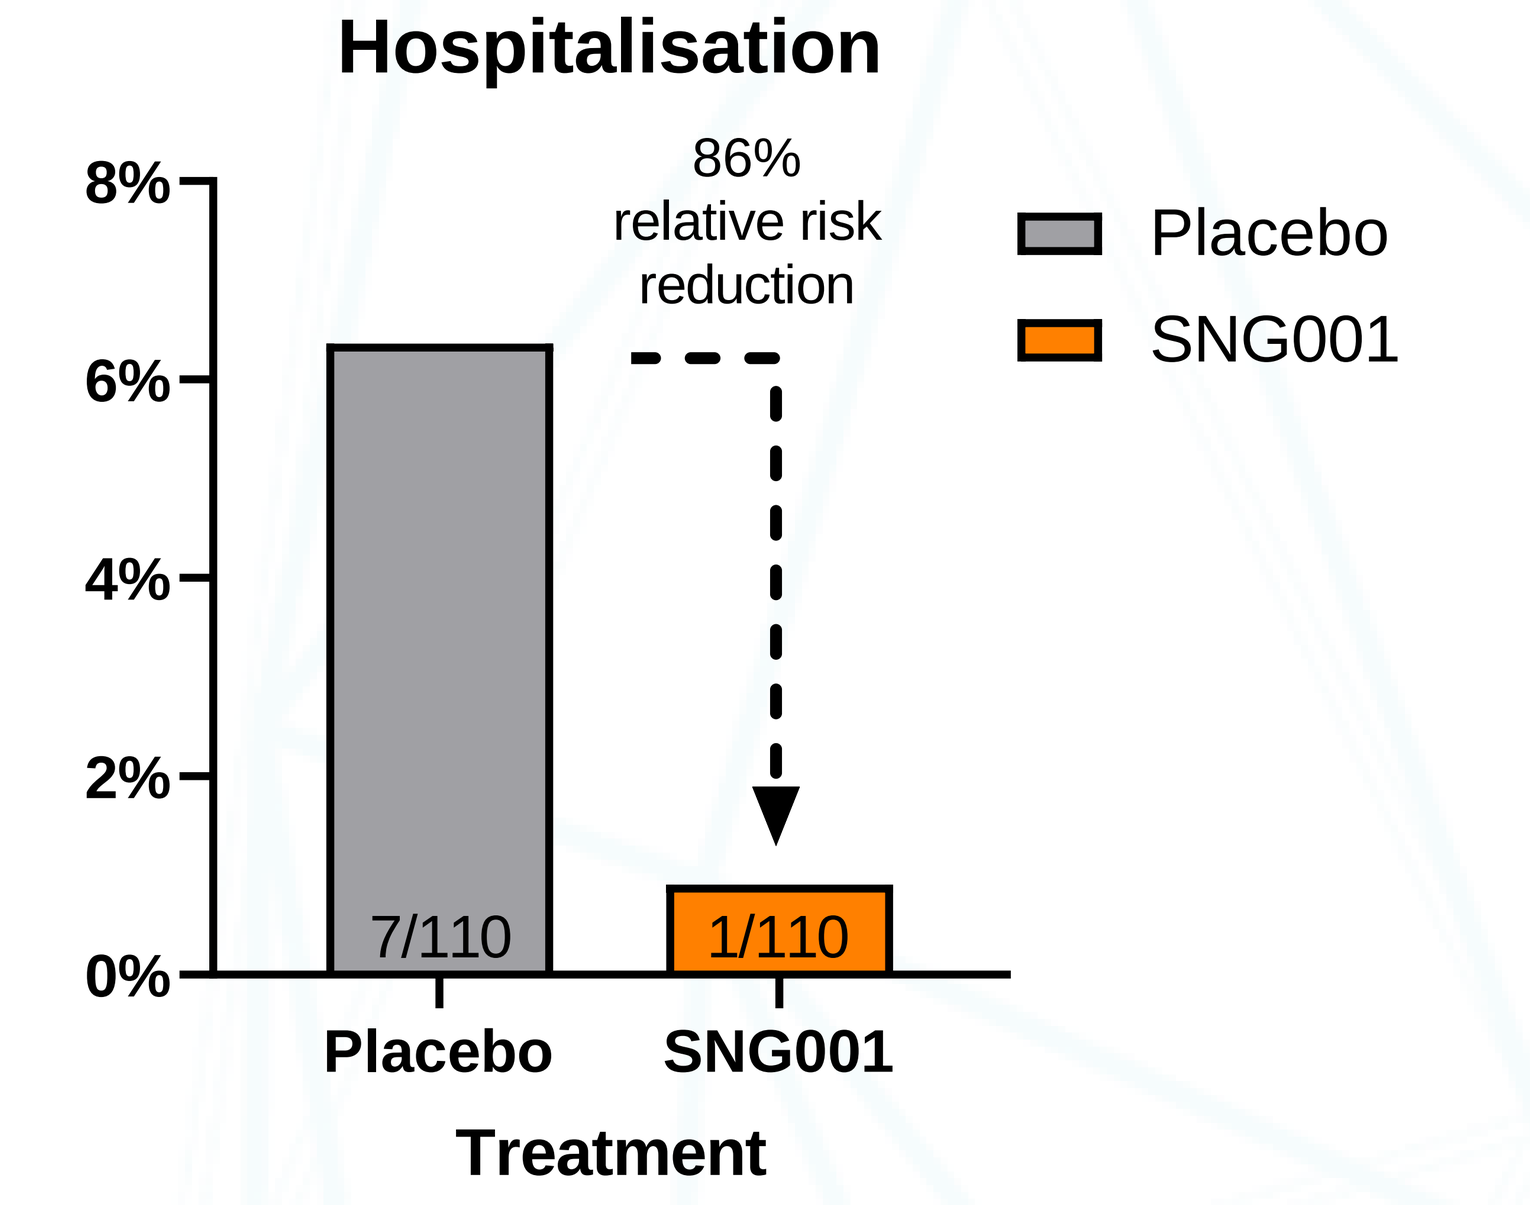
Hospitalisation
86% relative risk reduction
Placebo
SNG001
8%
6%
4%
2%
0%
7/110
1/110
Treatment

Acknowledgements

We would like to thank all the patients who have taken part in our clinical trials and the study teams who conducted them.

Disclosures

Synairgen is a UK-based biotech company developing inhaled interferon beta (SNG001) as a potential treatment for severe viral lung infections (www.Synairgen.com).